EXPRESSION PROFILES IN MATCHED PRIMARY AND RECURRENT OVARIAN CARCINOMAS

Deborah A. Zajchowski, PhD[1], Beth Y. Karlan, MD[2], and Laura K. Shawver, PhD[1]

[1]The Clearity Foundation, La Jolla, CA; [2]Cedars-Sinai Women's Cancer Program at SOCCI, Division of Gynecologic Oncology, Los Angeles, CA

INTRODUCTION

Little is known about the molecular characteristics of tumors present at relapse in advanced stage ovarian cancer patients following treatment with standard chemotherapy. Such knowledge could impact salvage therapy selection. Since 2008, we have profiled and maintained a database of biomarker expression results from tumor samples obtained from >125 ovarian cancer patients. These biomarkers include therapeutic targets as well as proteins correlated with response to chemotherapeutic drugs used in ovarian cancer treatment **(Table 1).**

Table 1. Biomarkers Evaluated in this Study with Associated Drug-Response Correlations

HUGO ID	NAME	MARKER EXPRESSION	DRUG RESPONSE
TOP1	Topoisomerase 1	High	topoisomerase 1 inhibitors
TOP2A	Topoisomerase 2A	High	topoisomerase 2 inhibitors
TS	Thymidylate synthase	Low	fluoropyrimidines
RRM1	Ribonucleotide reductase regulatory subunit M1	Low	gemcitabine
ERCC1	Excision repair complementation group 1	High	platinum resistance
PGP	P-glycoprotein, multi-drug resistance - 1	High	anthracycline/ taxane resistance
SPARC	Secreted protein, acidic, cysteine-rich	High	nab-paclitaxel
ESR1	Estrogen receptor	High	aromatase inhibitors
AR	Androgen receptor	High	anti-androgens
EGFR	Epidermal growth factor receptor	High	EGFR inhibitors
BCRP	Breast cancer resistance protein	High	topotecan/ mitoxanthrone resistance
COX-2	Cyclooxygenase-2	High	Cox-2 inhibitors
MRP1	Multidrug resistant protein 1		
MGMT	O-6-methyl guanine DNA methyltransferase	Low	temozolamide
Ki67	Proliferation antigen		

For marker references, visit http://www.clearityfoundation.org/drugs-and-biomarkers.aspx

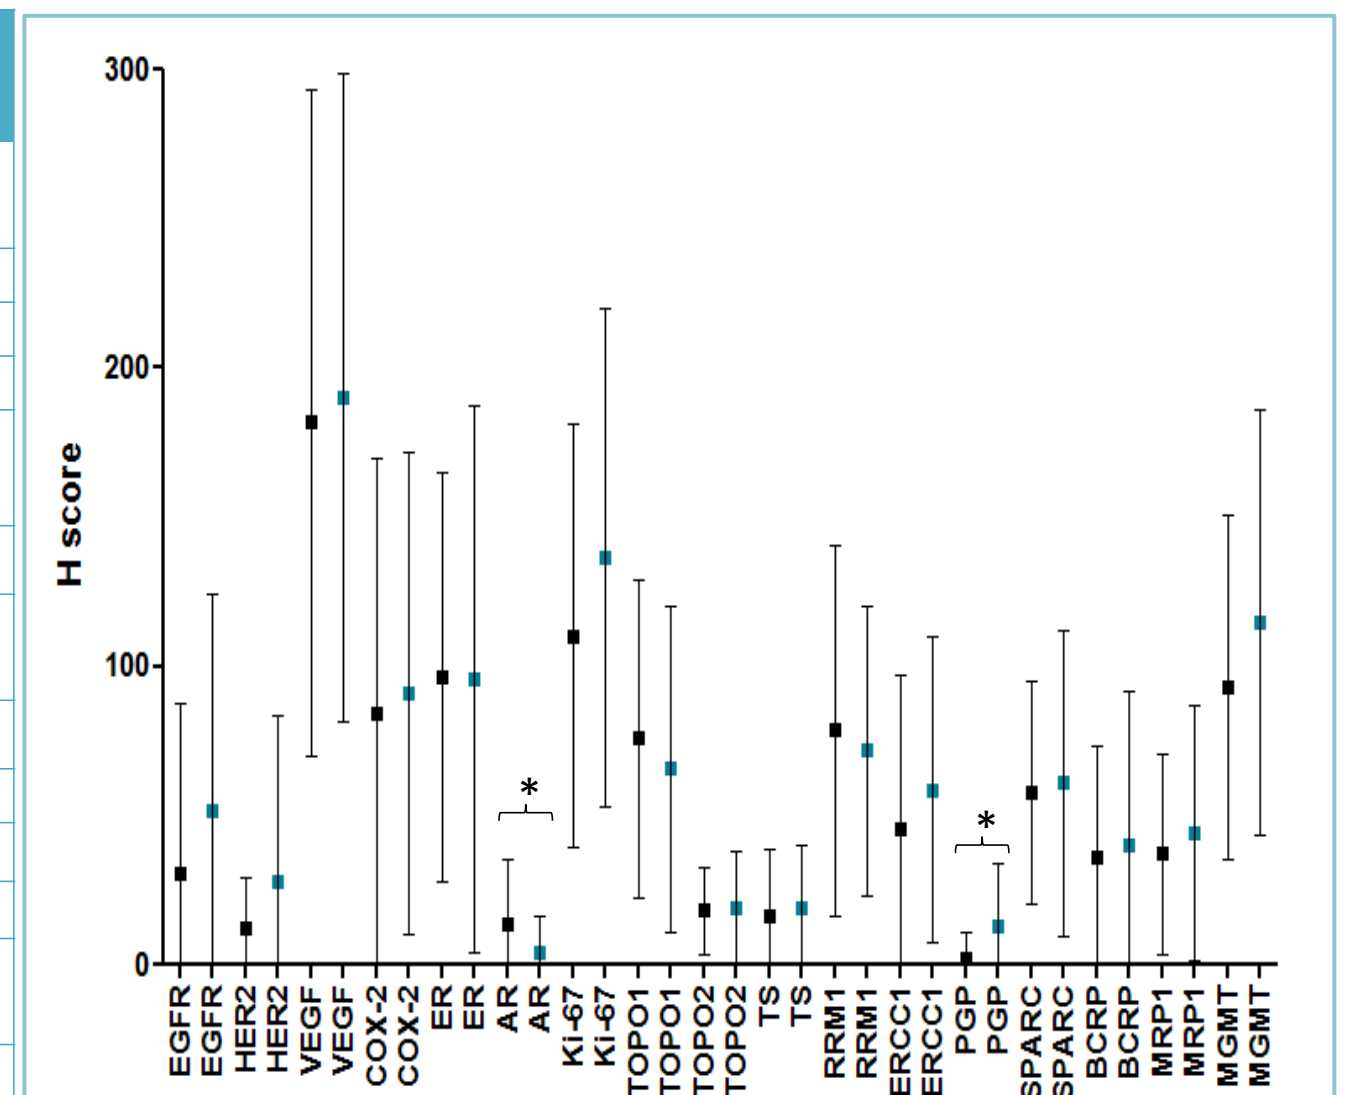
H score
300
200
100
0
EGFR
HER2
VEGF
COX-2
ER
AR
Ki-67
TOPO1
TOPO2
TS
RRM1
ERCC1
PGP
SPARC
BCRP
MRP1
MGMT
*

Figure 1. Biomarker Expression in Primary and Recurrent Ovarian Serous Carcinoma and Adenocarcinoma Specimens Expression data graphed as H scores for indicated proteins. Squares, mean ± SD for 70 primary (black) and 48 recurrent (teal) specimens are tabulated. *, $p < 0.05$, students' ttest

Our analysis of data from 118 primary and recurrent ovarian serous carcinoma samples within this database revealed that the average expression of PGP (MDR1) and AR were significantly different in these two groups **(Figure 1).** Increased expression of PGP and decreased levels of AR were observed in recurrent specimens.

To determine whether these or other differences between recurrent and primary lesions would be observed if the corresponding specimens from the same patient were analyzed, we performed an analysis of matched pairs of samples.

MATERIALS and METHODS

Patients and tumor specimens. The patients are an unselected population who sought molecular profiling assistance from The Clearity Foundation between October, 2008 and May, 2011. Formalin-fixed, paraffin-embedded (FFPE) tissue specimens and patient treatment histories were obtained under written informed consent. Specimens procured during primary surgical procedures were from ovary and fallopian tube (O) or the peritoneal cavity (P; biopsies from omentum, diaphragm, peritoneum, colon, appendix, cul de sac, side wall). Recurrent cancer specimens were from the peritoneal cavity (M), lymph nodes (LN), or distant organs (MD; lung, liver).

Immunohistochemistry. CLIA-certified laboratories have performed these analyses to ensure that the protocols and reagents used have been fully validated as well as high reproducibility of the test results. Immunohistochemistry (IHC) analysis of ER, PTEN, MDR1, BCRP, MRP1, ERCC1, MGMT, TS, RRM1, TOP1/2A was performed by Caris Life Sciences, Inc using the Ventana or the DAKO automated staining systems. EGFR, VEGF, PTEN, COX2, and Ki67 IHC analyses were performed at Clarient, Inc. Following heat-induced epitope retrieval, antibody incubation was for 20-40 minutes (antibody-specific), visualization was by the Ultraview or Vision Biosystem Novolink Poly-HRP (Ventana) or Biocare envision plus horseradish peroxidase Polymer Detection System (DAKO). All slides were scored manually by a board certified pathologist and results reported as % of tumor cells that stained positive and intensity of staining (0, 1+, 2+, 3+). Expression data are presented as histoscores- product of % and intensity.

Expression of the biomarker panel was evaluated in 38 matched primary and recurrent specimens from 17 ovarian cancer patients (**Table 2).**

Table 2. Characteristics of Patients and Samples Profiled in this Study

ID #	Histology	Stage	Platinum response	Specimen Tissue Site			
				Primary	Recurrences		
10	Serous	IIIB	Sensitive	Ovary		Pleural Fluid	
11	Serous	III	Sensitive	Ovary/ Fallopian Tube	Omentum	Peritoneal Fluid	
18	Clear Cell	IIIC	Refractory	Ovary/ Fallopian Tube	Abdomen		
26	Serous	IV	Sensitive	Ovary		Liver	Liver
27	Serous	IIIC	Resistant	Omentum		Peritoneal Fluid	
29	Serous	IIIC	Sensitive		Lymph Node	Pleural Fluid	Pleural Fluid
34	Serous	IIIC	Sensitive	Ovary/ Fallopian Tube	Omentum		
35	Serous-endometrioid	IIIC	Sensitive	Ovary	Connective and Soft tissue		
42	Serous	IIIB	Sensitive	Omentum	Omentum	Breast	
45	Serous	IIIC	Sensitive		Cul-de-Sac	Side wall	
72	Adenocarcinoma	IIIC	Sensitive		Connective and Soft tissue	Connective and Soft tissue	
73	Serous	IIIC	Sensitive	Ovary	Lymph Node		
75	Serous	III	Sensitive	Ovary	Pelvis	Pleural Fluid	
95	Serous	IIIC	Resistant	Ovary/ Fallopian Tube	Pelvis	Pleural Fluid	
96	Endometrioid	IC	Sensitive	Ovary	Colon	Omen/Pelvis	
116	Endometrioid	IIIC	Sensitive		Peritoneum	Pelvis	

Similar expression of MGMT, ER, HER2, and VEGF were observed in patient-matched specimens.
ERCC1, BCRP, EGFR, TS, and PGP levels differed by ≥4-fold in more than 45% of the paired specimens **(Figure 2).**

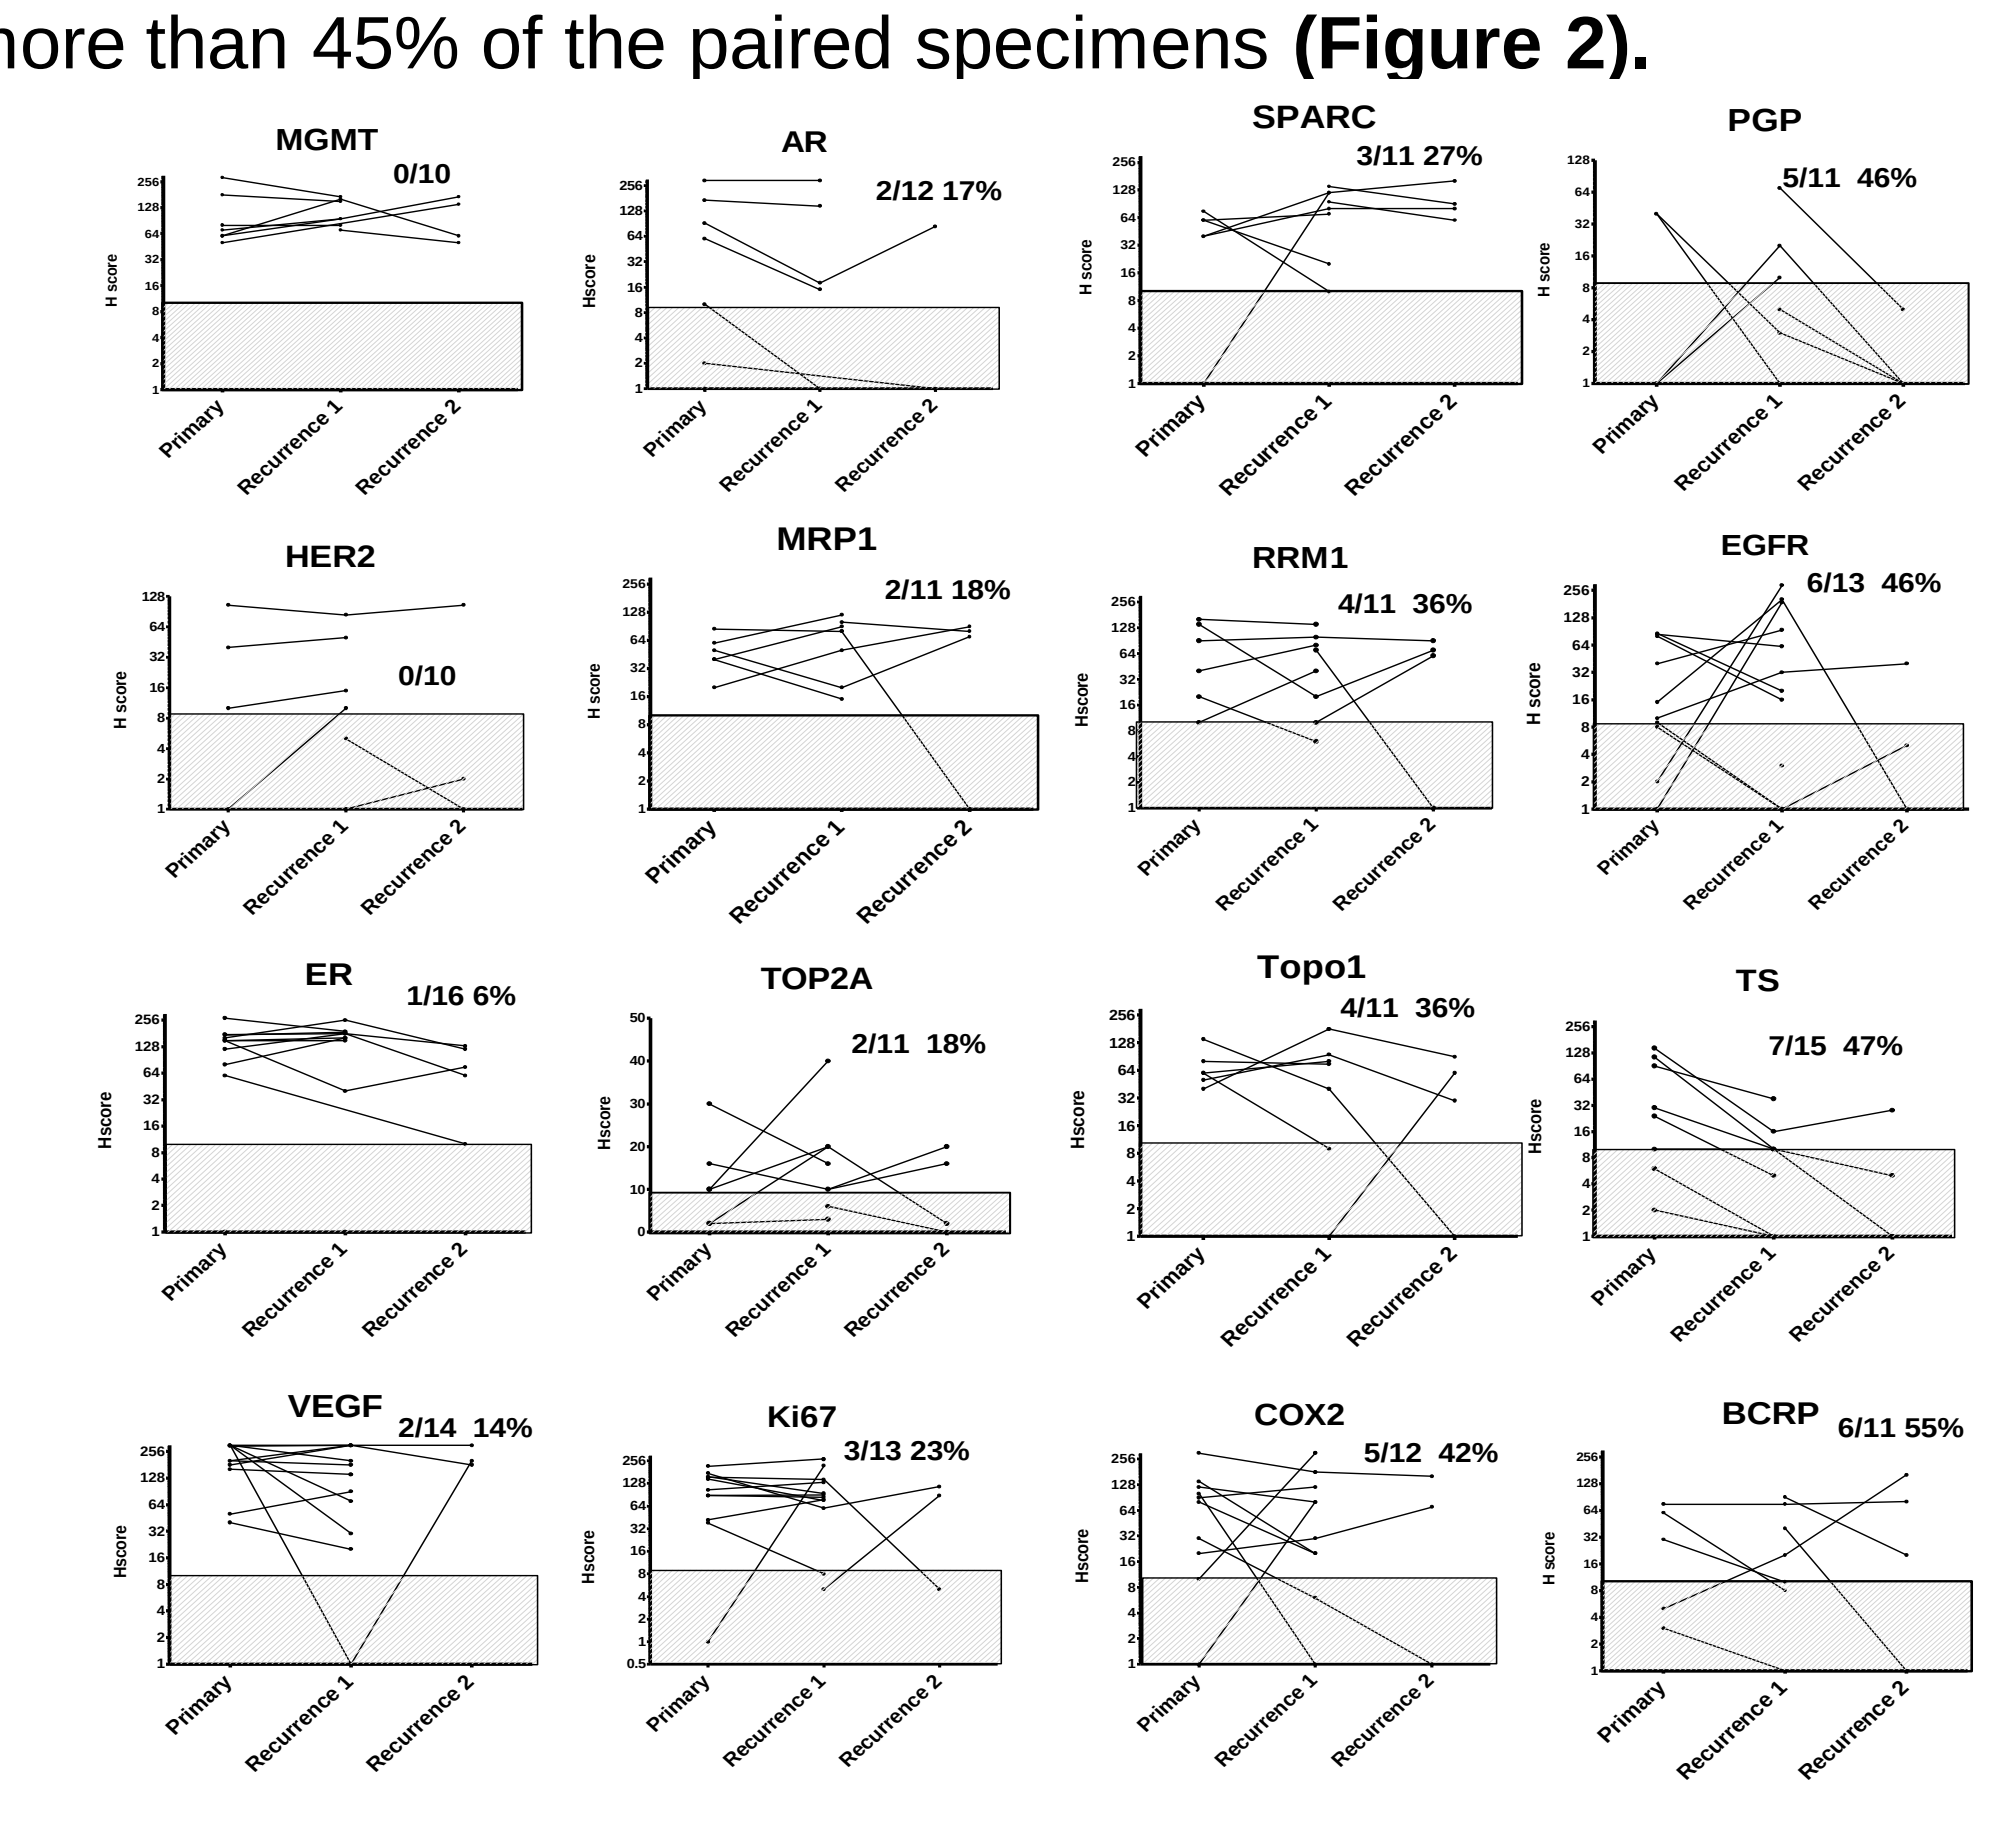
MGMT 0/10
AR 2/12 17%
SPARC 3/11 27%
PGP 5/11 46%
HER2 0/10
MRP1 2/11 18%
RRM1 4/11 36%
EGFR 6/13 46%
ER 1/16 6%
TOP2A 2/11 18%
Topo1 4/11 36%
TS 7/15 47%
VEGF 2/14 14%
Ki67 3/13 23%
COX2 5/12 42%
BCRP 6/11 55%
H score
Primary
Recurrence 1
Recurrence 2

Figure 2. Expression of biomarker panel in matched primary and recurrent specimens. Protein expression levels for primary and matched recurrences (1 and 2) are connected by lines and graphically represented as H scores. Fraction, number of specimens with ≥4-fold variation in expression from matched sample/total # pairs; paired samples with H scores ≤10 (inside hatched box) were not considered in this measurement.

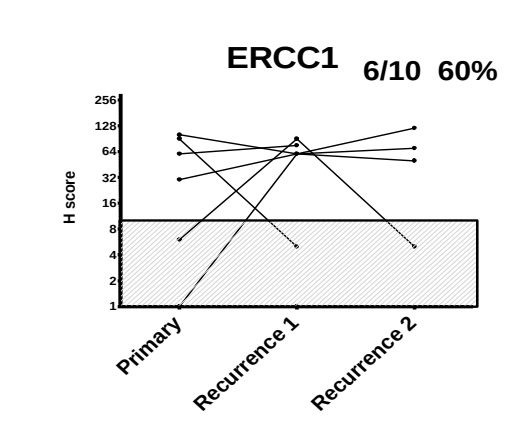
ERCC1 6/10 60%
H score
Primary
Recurrence 1
Recurrence 2

Similar expression of multiple markers observed in most matched samples (see 96E, 35SE, and 116E; **Figure 3)**. But, single markers are differentially expressed.

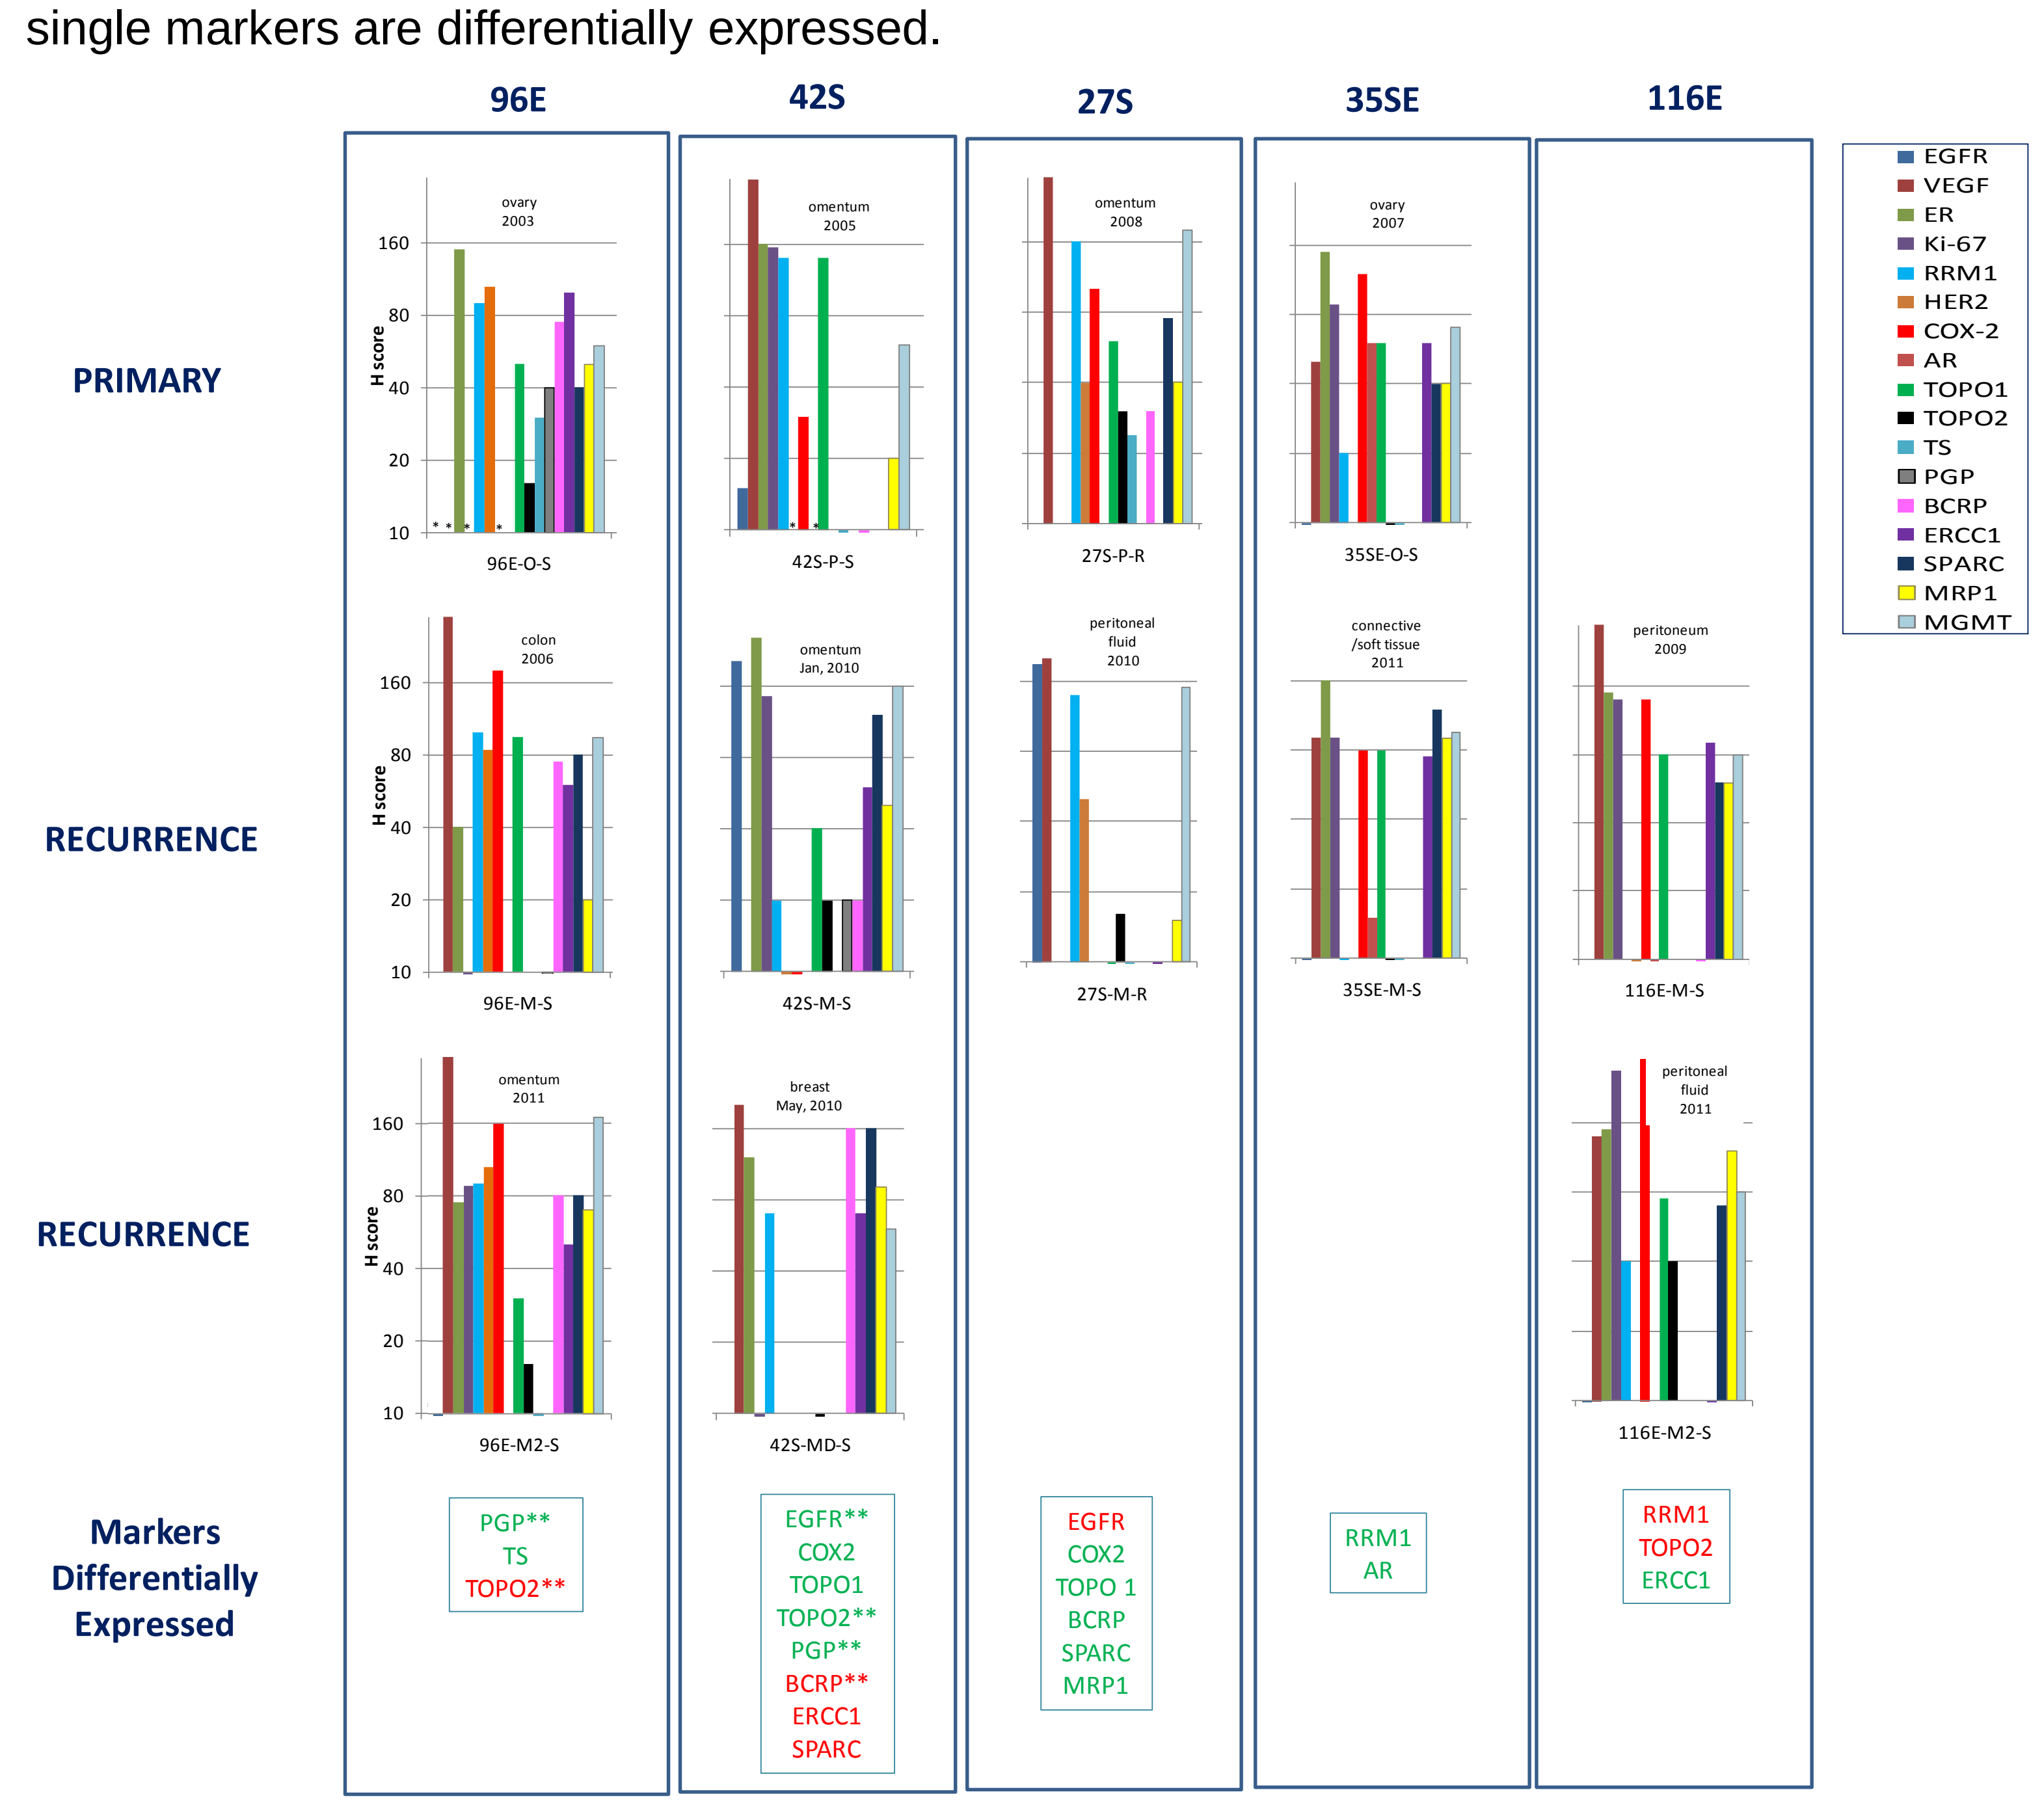
96E
42S
27S
35SE
116E
PRIMARY
RECURRENCE
RECURRENCE
Markers Differentially Expressed
96E-O-S
42S-P-S
27S-P-R
35SE-O-S
96E-M-S
42S-M-S
27S-M-R
35SE-M-S
116E-M-S
96E-M2-S
42S-MD-S
116E-M2-S
PGP**
TS
TOPO2**
EGFR**
COX2
TOPO1
TOPO2**
PGP**
BCRP**
ERCC1
SPARC
EGFR
COX2
TOPO 1
BCRP
SPARC
MRP1
RRM1
AR
RRM1
TOPO2
ERCC1
EGFR
VEGF
ER
Ki-67
RRM1
HER2
COX-2
AR
TOPO1
TOPO2
TS
PGP
BCRP
ERCC1
SPARC
MRP1
MGMT

Figure 3. Comparison of marker profiles in matched specimens from five patients (indicated at top). Marker expression (H scores) graphically represented for each specimen with tissue source and date above each graph. Differentially expressed markers listed below each matched set. red, up-regulated and green, down-regulated in most recent specimen.
*, Not tested; **, markers expressed in one recurrence. Scale, Log 2; values < 10 not shown.

- Drug resistance proteins /transporters BCRP and PGP/MDR1 were differentially expressed and reduced in expression in later recurrences from the same patient (see 96E, 42S).
- ERCC1 expression variable in recurrent lesions (see 42S and 116E)
- Highly elevated EGFR levels in two recurrences (see 42S and 27S).
- Discordant expression of EGFR, VEGF, Topo1, and BCRP in metachronous recurrences in omentum and breast (42S). These proteins are expressed in the primary tumor, suggesting existence in the primary tumor of heterogeneous cell populations with differing tropism for metastatic sites.

Do the differences in marker expression measured in recurrences have clinical importance?
Would these differences impact choice of therapy if biomarkers are utilized?

We modeled these data using expression cut-points for selecting treatment with chemotherapeutic drugs based upon clinical research evidence and our database of 125 ovarian tumor profiles **(Figure 4).**

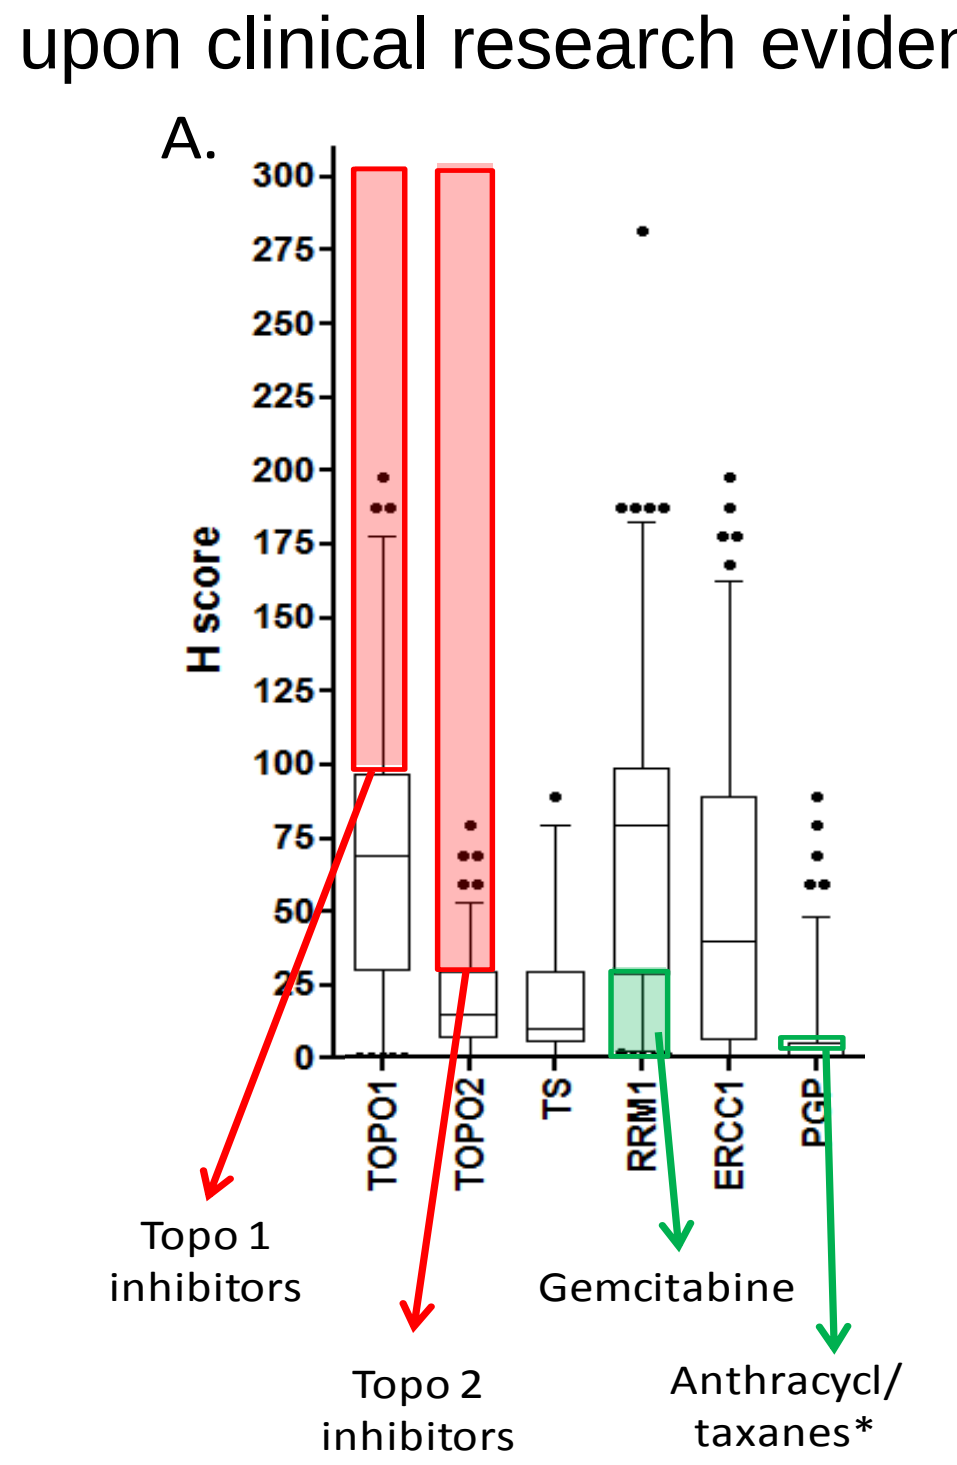
A.
H score
TOPO1
TOPO2
TS
RRM1
ERCC1
PGP
Topo 1 inhibitors
Topo 2 inhibitors
Gemcitabine
Anthracycl/ taxanes*

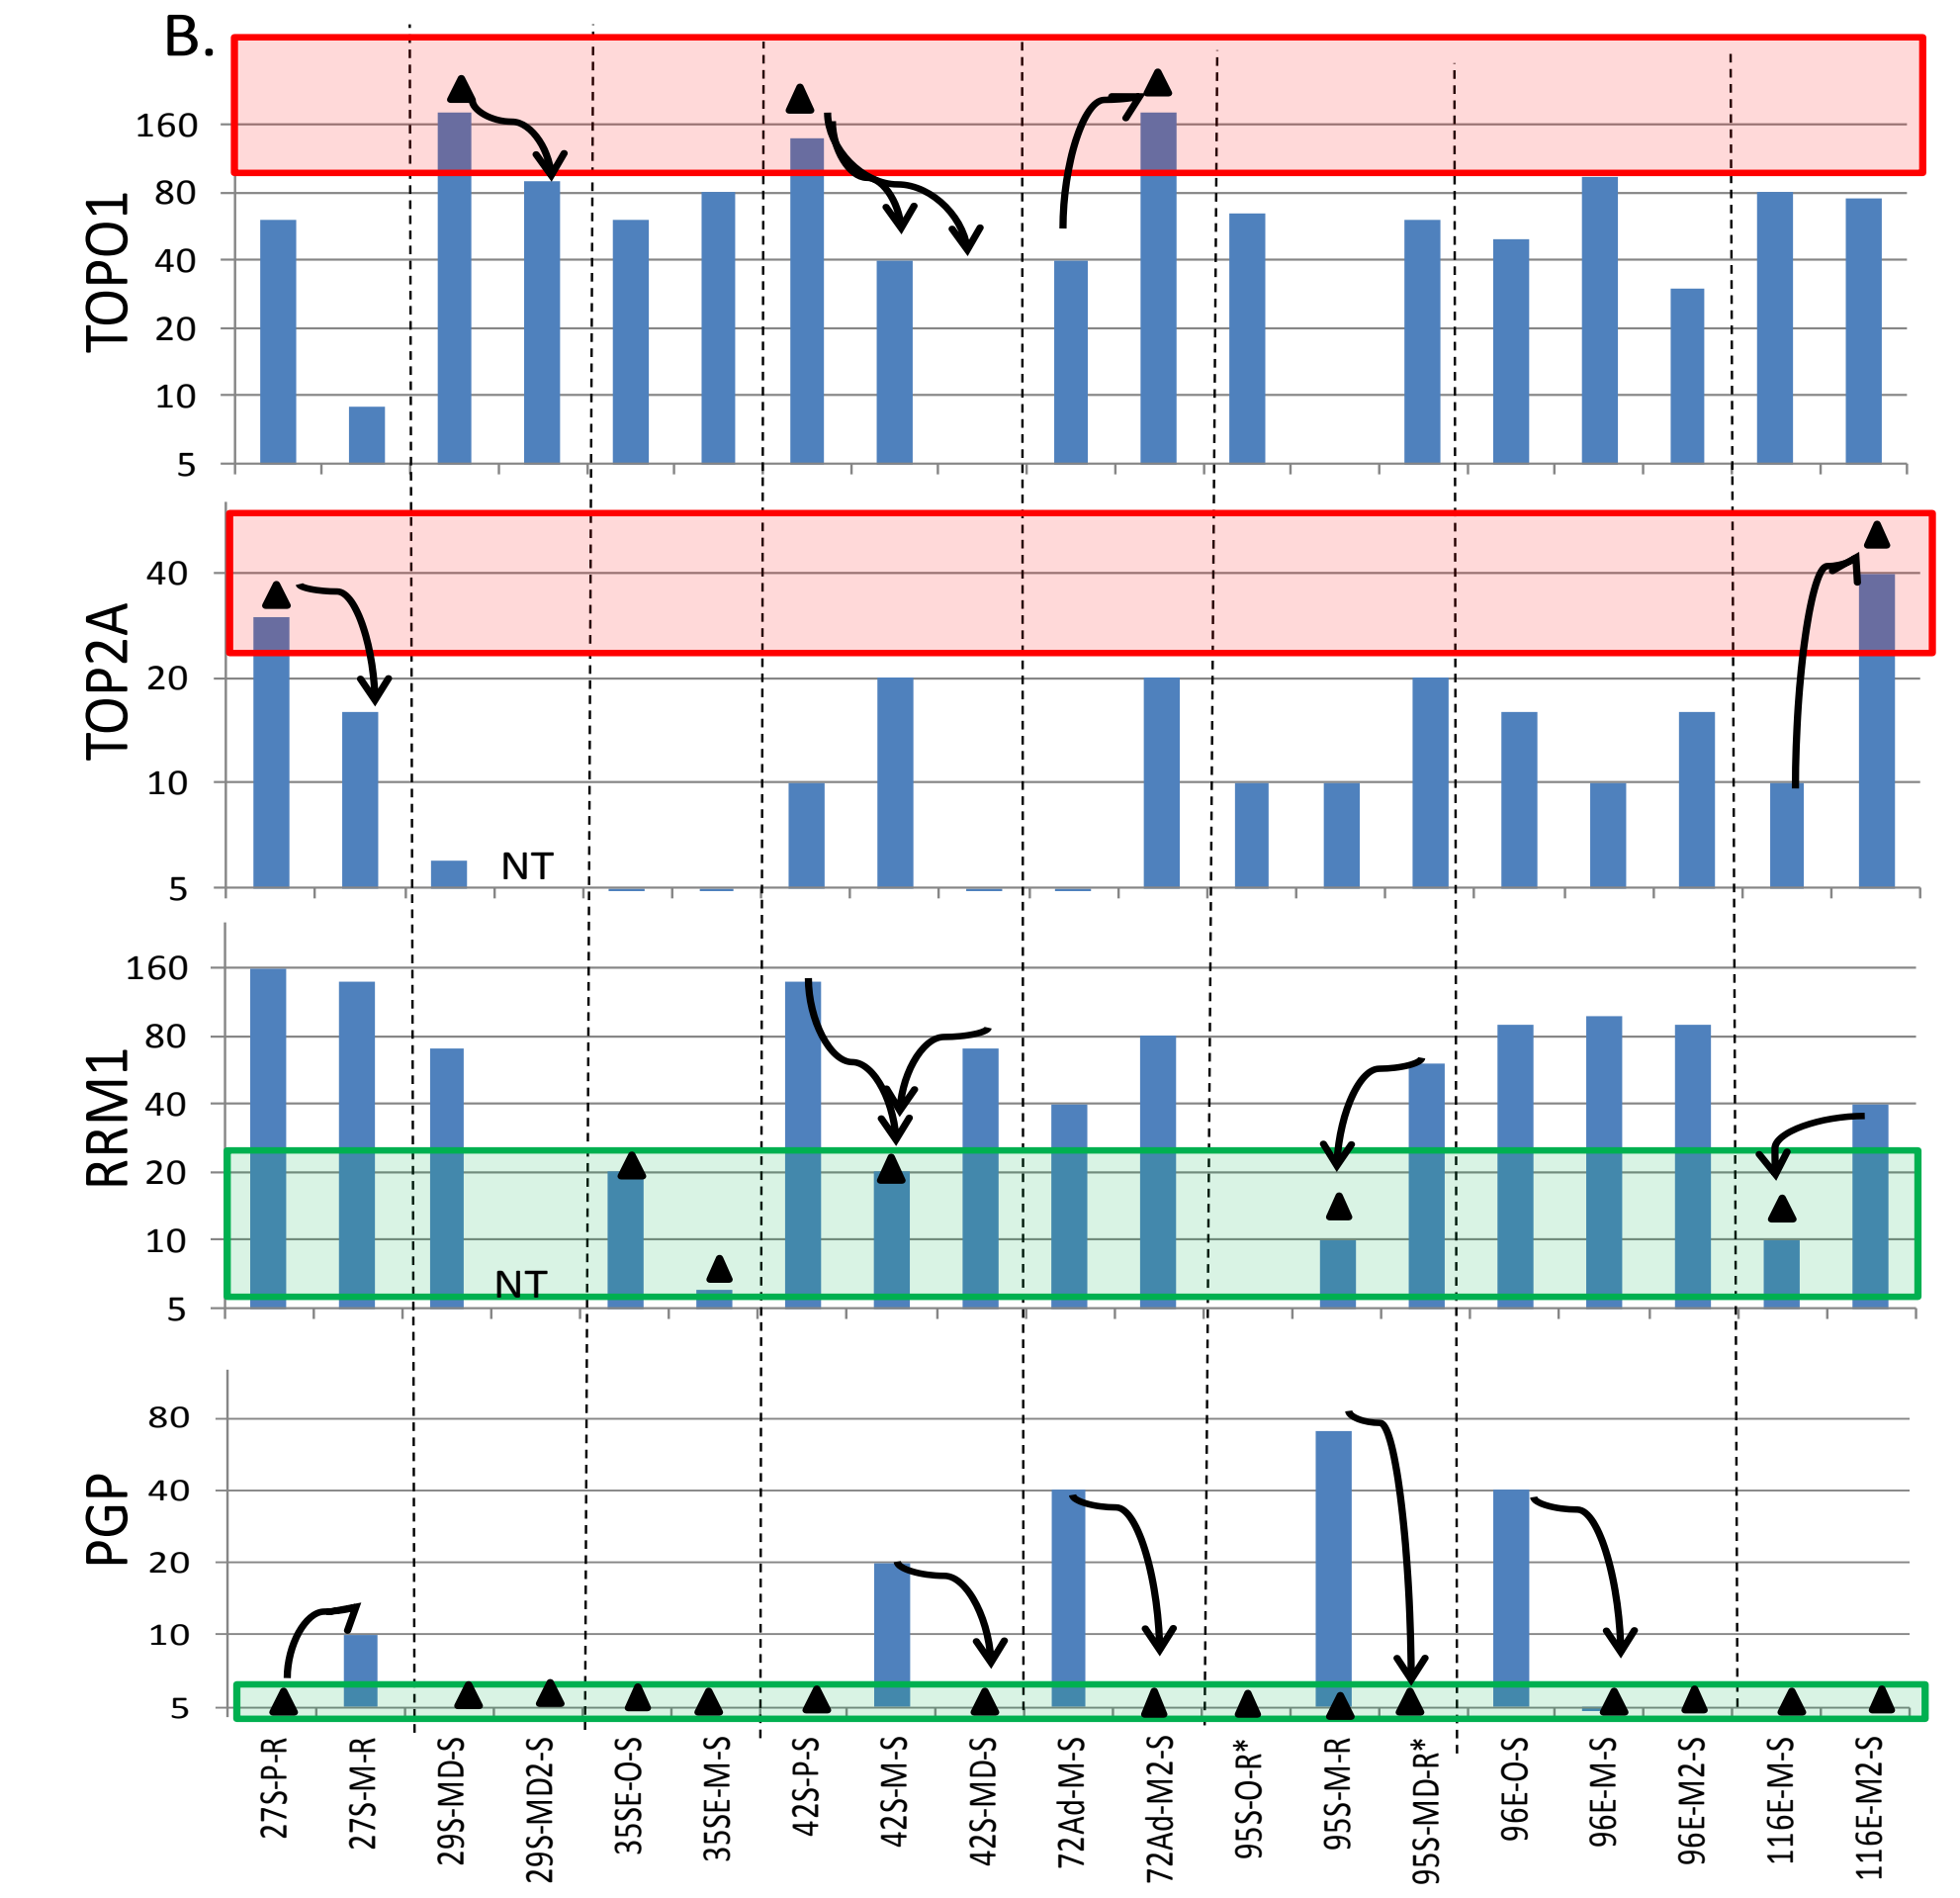
B.
TOPO1
TOP2A
RRM1
PGP
NT
27S-P-R
27S-M-R
29S-MD-S
29S-MD2-S
35SE-O-S
35SE-M-S
42S-P-S
42S-M-S
42S-MD-S
72Ad-M-S
72Ad-M2-S
95S-O-R*
95S-M-R
95S-MD-R*
96E-O-S
96E-M-S
96E-M2-S
116E-M-S
116E-M2-S

Clinical impact of marker expression variation can be significant if the marker expression crosses a cut-point that determines therapy selection.

Figure 4. Modeling of therapy selection based on biomarker expression in patient-matched primary and recurrent samples. A, Rectangles designate marker expression levels that trigger assignment of indicated therapies based on top 25th (red) or bottom 25th percentile (green) of tumors profiled (see Figure 1); positive expression of PGP supports use of anthracyclines or taxanes in concert with other markers. B, Expression data as H scores for indicated markers in matched samples. Rectangles designate expression levels that trigger assignment of therapy (see A); black triangles, expression result supports therapy assignment; arrow, designates results in recurrent lesion that would change decision for therapy selection. P, primary; M, recurrence; MD, distant recurrence

SUMMARY and CONCLUSIONS

Analysis of 118 primary and recurrent lesions obtained from serous ovarian cancer patients revealed significant differences in the expression of key biomarkers that are associated with treatment response.

Comparison of expression of these biomarkers in patient-matched primary and recurrent samples revealed that MGMT, ER, HER2, and VEGF were very similar in matched specimens, whereas expression of ERCC1, BCRP, EGFR, TS, and PGP were frequently discordant in specimens from the same patient.

Influence of sample acquisition, processing, and storage was not measured in this study but could impact immuno-reactivity for some of these antigens and lead to differences in the measured expression levels. While sample collection needs to be standardized, changes due to biological variation are likely.

Recurrent lesions have different expression profiles of drug target and resistance markers from their corresponding primary lesions and from prior recurrences. These differences may be sufficiently large as to impact selection of therapy. Therefore, proximal tumor specimens are needed in order to have the most relevance for potential treatment choices.

These pilot data must be validated in a larger study, and if confirmed, support the need for profiling recurrent tumor specimens rather than archival blocks in future clinical trials.

ACKNOWLEDGEMENTS

We thank the patients and physicians who have participated in this study and are also appreciative of the efforts of Michelle Bernabe and Brenda Durand for patient support services, Kathleen Zajchowski for database coordination, and Michael Petka for enhancement of our database capabilities.